Abstract #2589

In vitro cytotoxic activities of the membrane active nonapeptide LTX-315 (Oncopore™) against human melanoma cells

LIV-MARIE EIKE[1], NANNAN YANG[1], ØYSTEIN REKDAL[1,2], BALDUR SVEINBJØRNSSON [1,2]
[1] Dept of Molecular Inflammation Research, Institute of Medical Biology, Faculty of Health, University of Tromsø,N-9037
[2] Lytix Biopharma, 9294, Tromsø, Norway

Lytix Biopharma

Background

Cationic antimicrobial peptides (CAPs) are a diverse group of highly conserved peptides found in virtually all species of life as a part of the innate immune system. In addition to the bactericidal effects, CAPs have been shown to have antitumor activity[1-4].

LTX-315 is an anti-cancer nonamer peptide derived from bovine lactoferricin (Fig 1). Experimental studies in animal models have shown that intralesional treatment with LTX-315 induces complete regression and systemic protective immune responses [5]. Tumor cells treated with LTX-315 are rapidly killed by a lytic mode of action with the potential to initiate inflammation. A phase 1/2a study is ongoing with LTX-315.

Aim

To investigate the mode of action underlying the cytotoxic activity of LTX-315 against a human melanoma cell line

LTX-315

Fig. 1 - Chemical structure of LTX-315

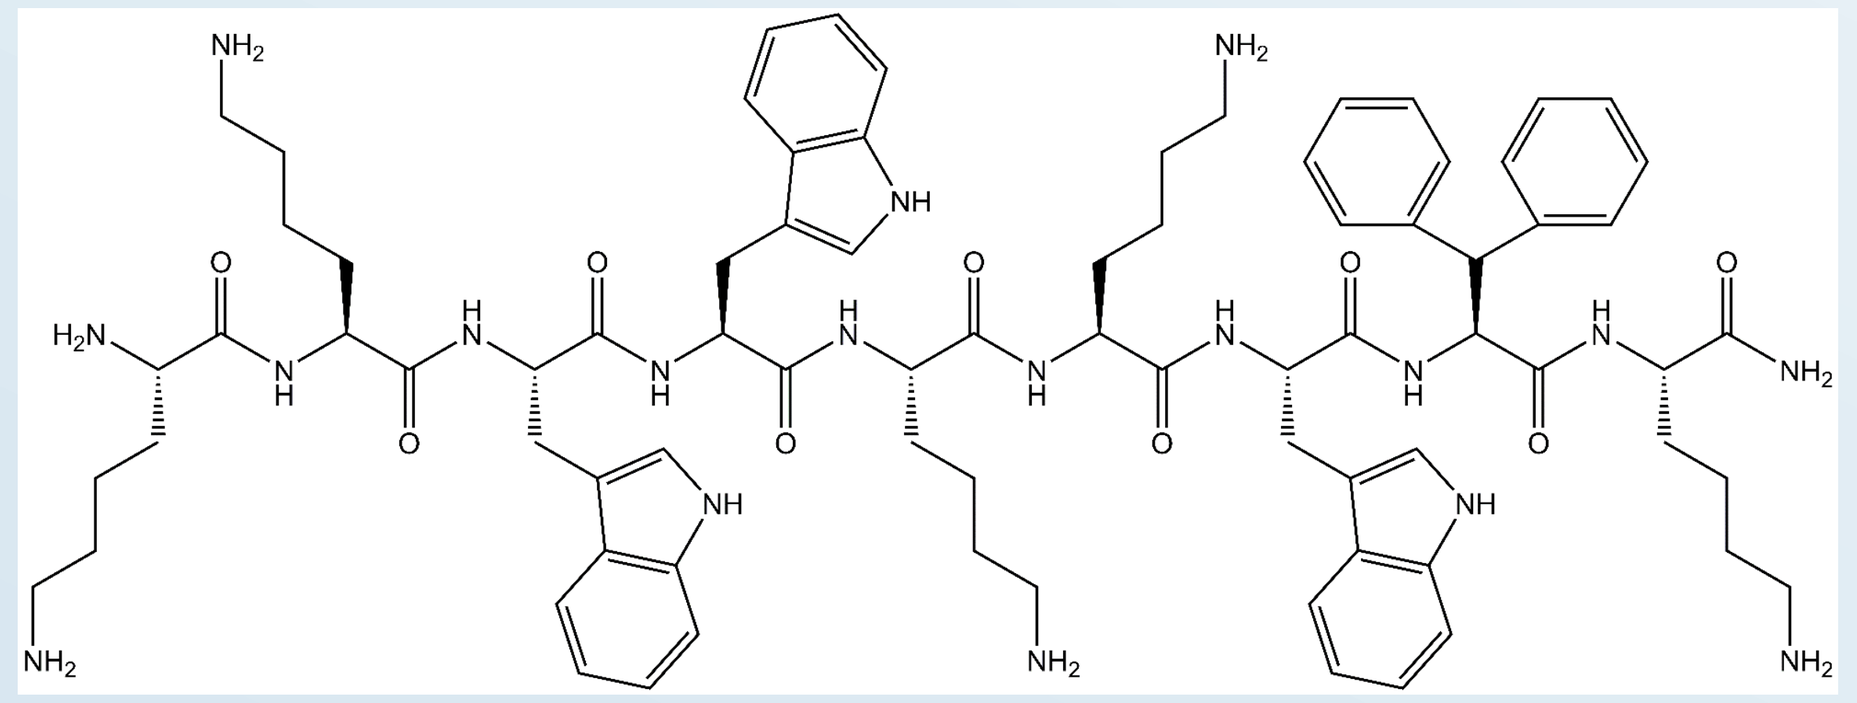

Results

Fig. 2 - In vitro kinetics of LTX-315

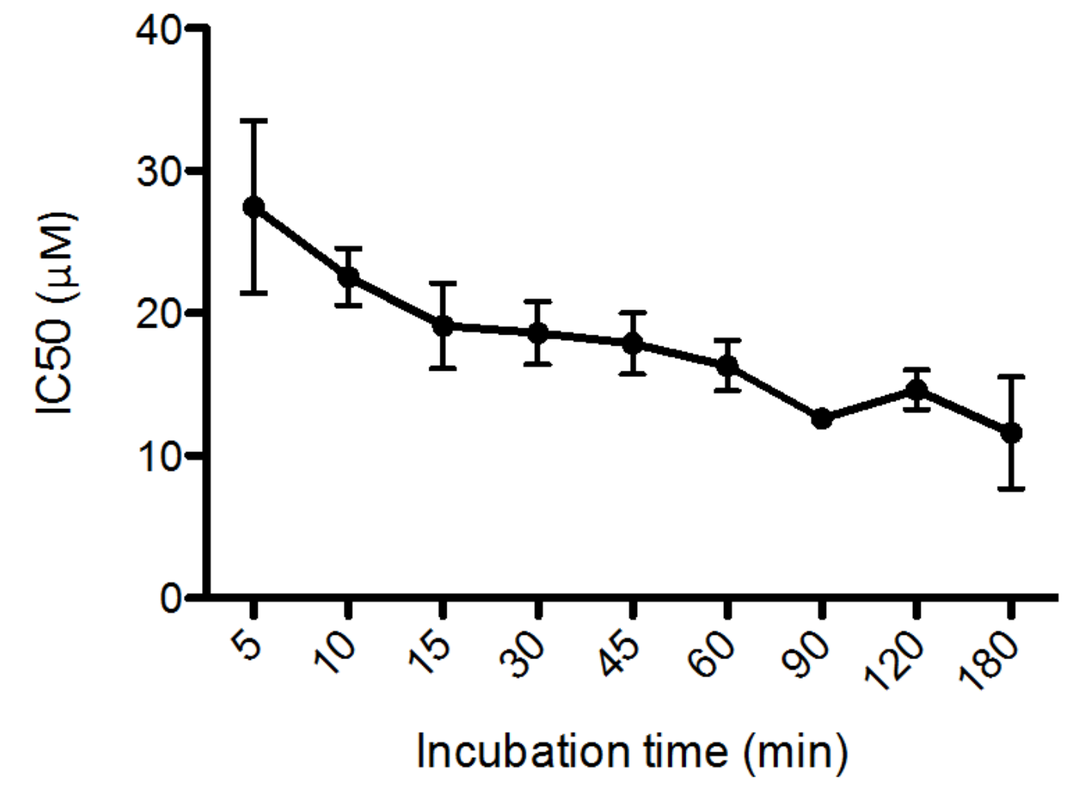
IC50 (µM)
40
30
20
10
0
5
10
15
30
45
60
90
120
180
Incubation time (min)

In vitro cell killing kinetics of LTX-315 against human melanoma cell line A375 measured by MTT. Result from three experiments are presented for each timepoint as mean ± SD.

Fig. 3 - The cancer cells are killed by a lytic mode of action

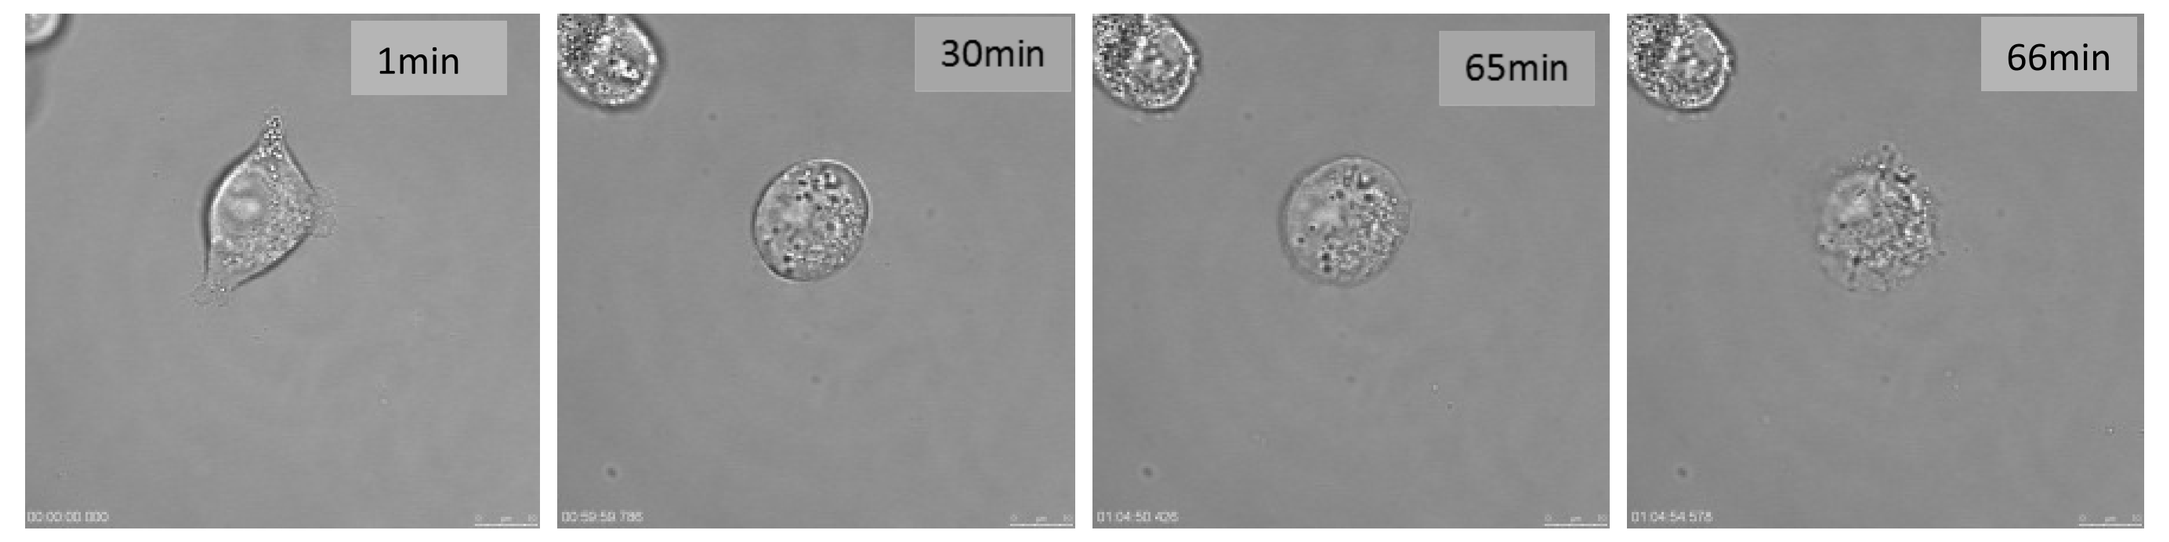
1min
30min
65min
66min

Bright field confocal images of A375 cells treated with 17 µM LTX-315.

Fig. 4 - LTX-315 internalizes and associates with mitochondria.

At low concentration, fluorescence-labeled peptide was associated with mitochondria (Fig 4) with a subsequent disintegration of the mitochondria membrane (Fig 5) also shown at ultrastructural level (Fig 6).

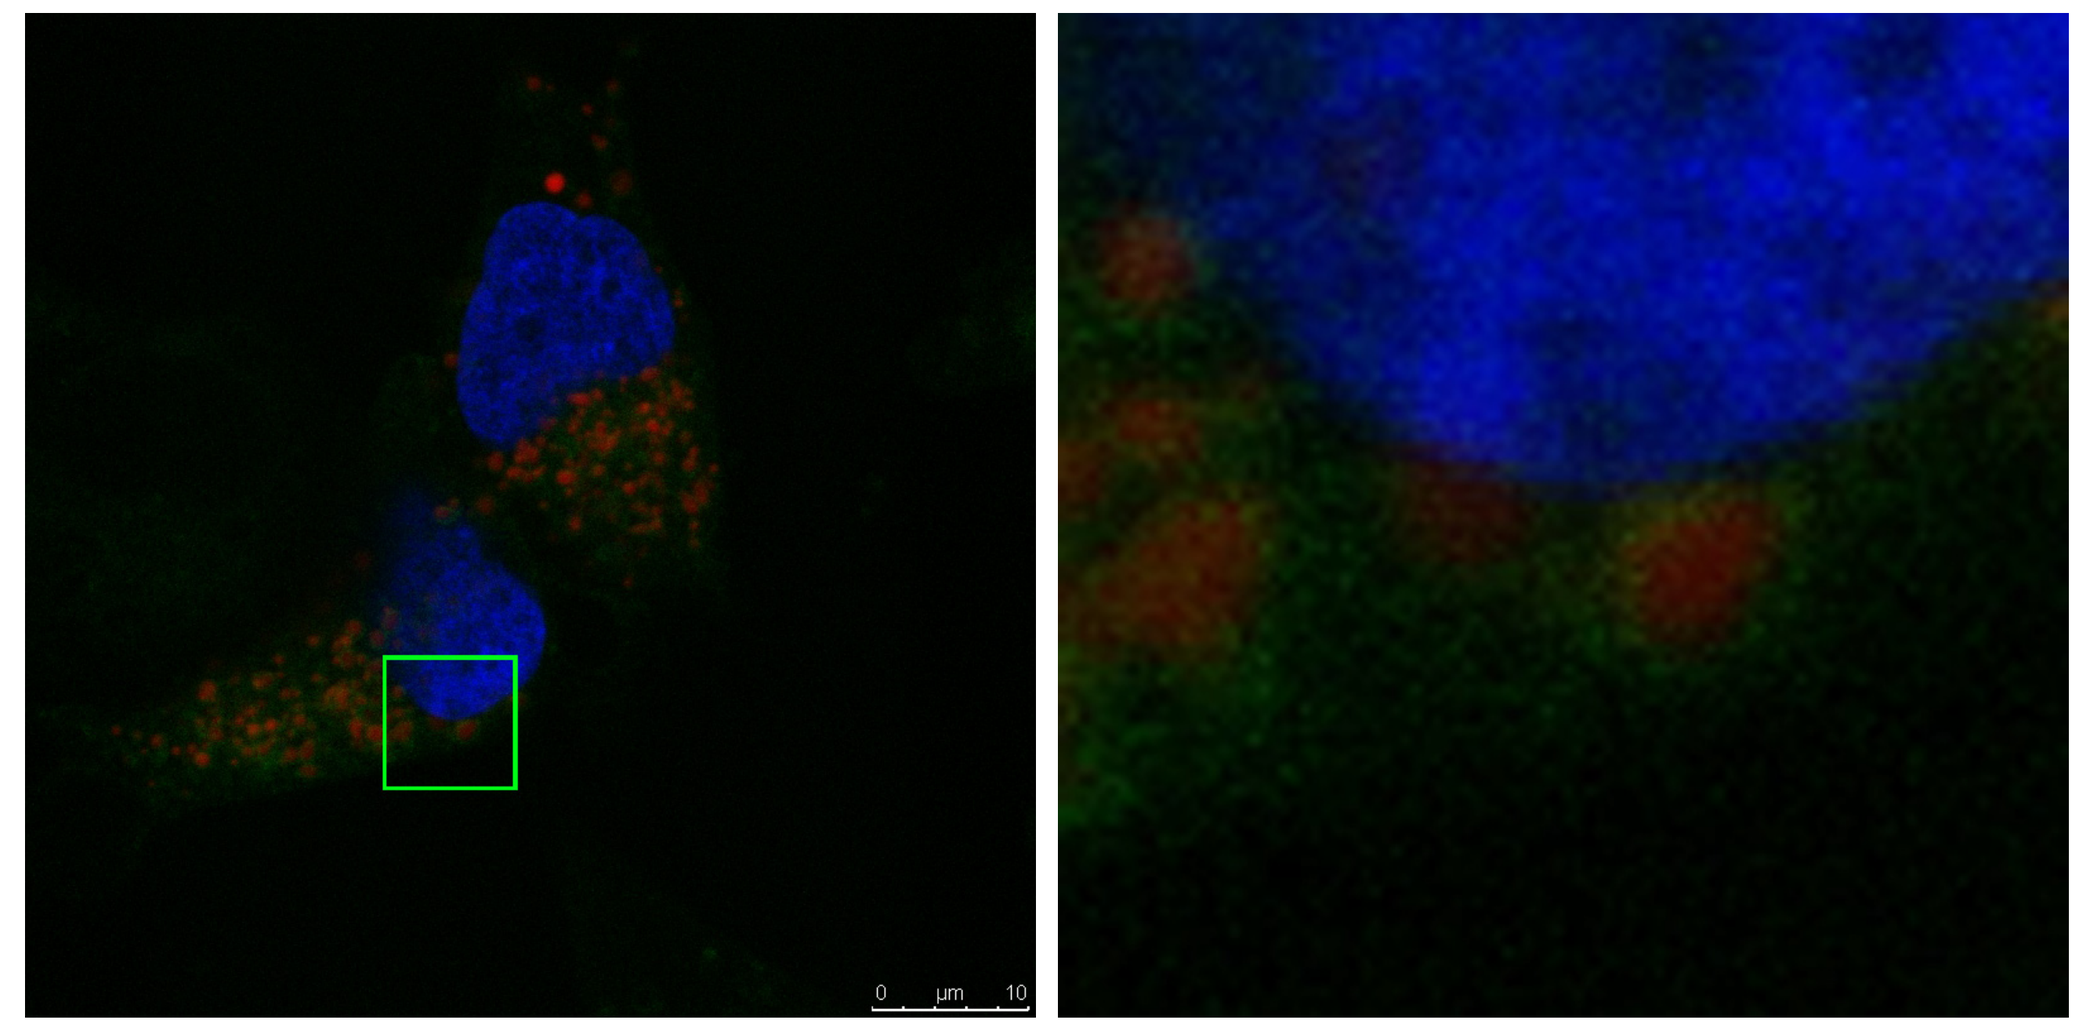

A375 cells treated with 3 µM LTX-315 for 30min and investigated with confocal microscopy. Fluorescense labeled LTX-315 (green), and labeled mitochondria (red) and nucleus (blue).

Fig. 5 - LTX-315 induces disintegration of mitochondria

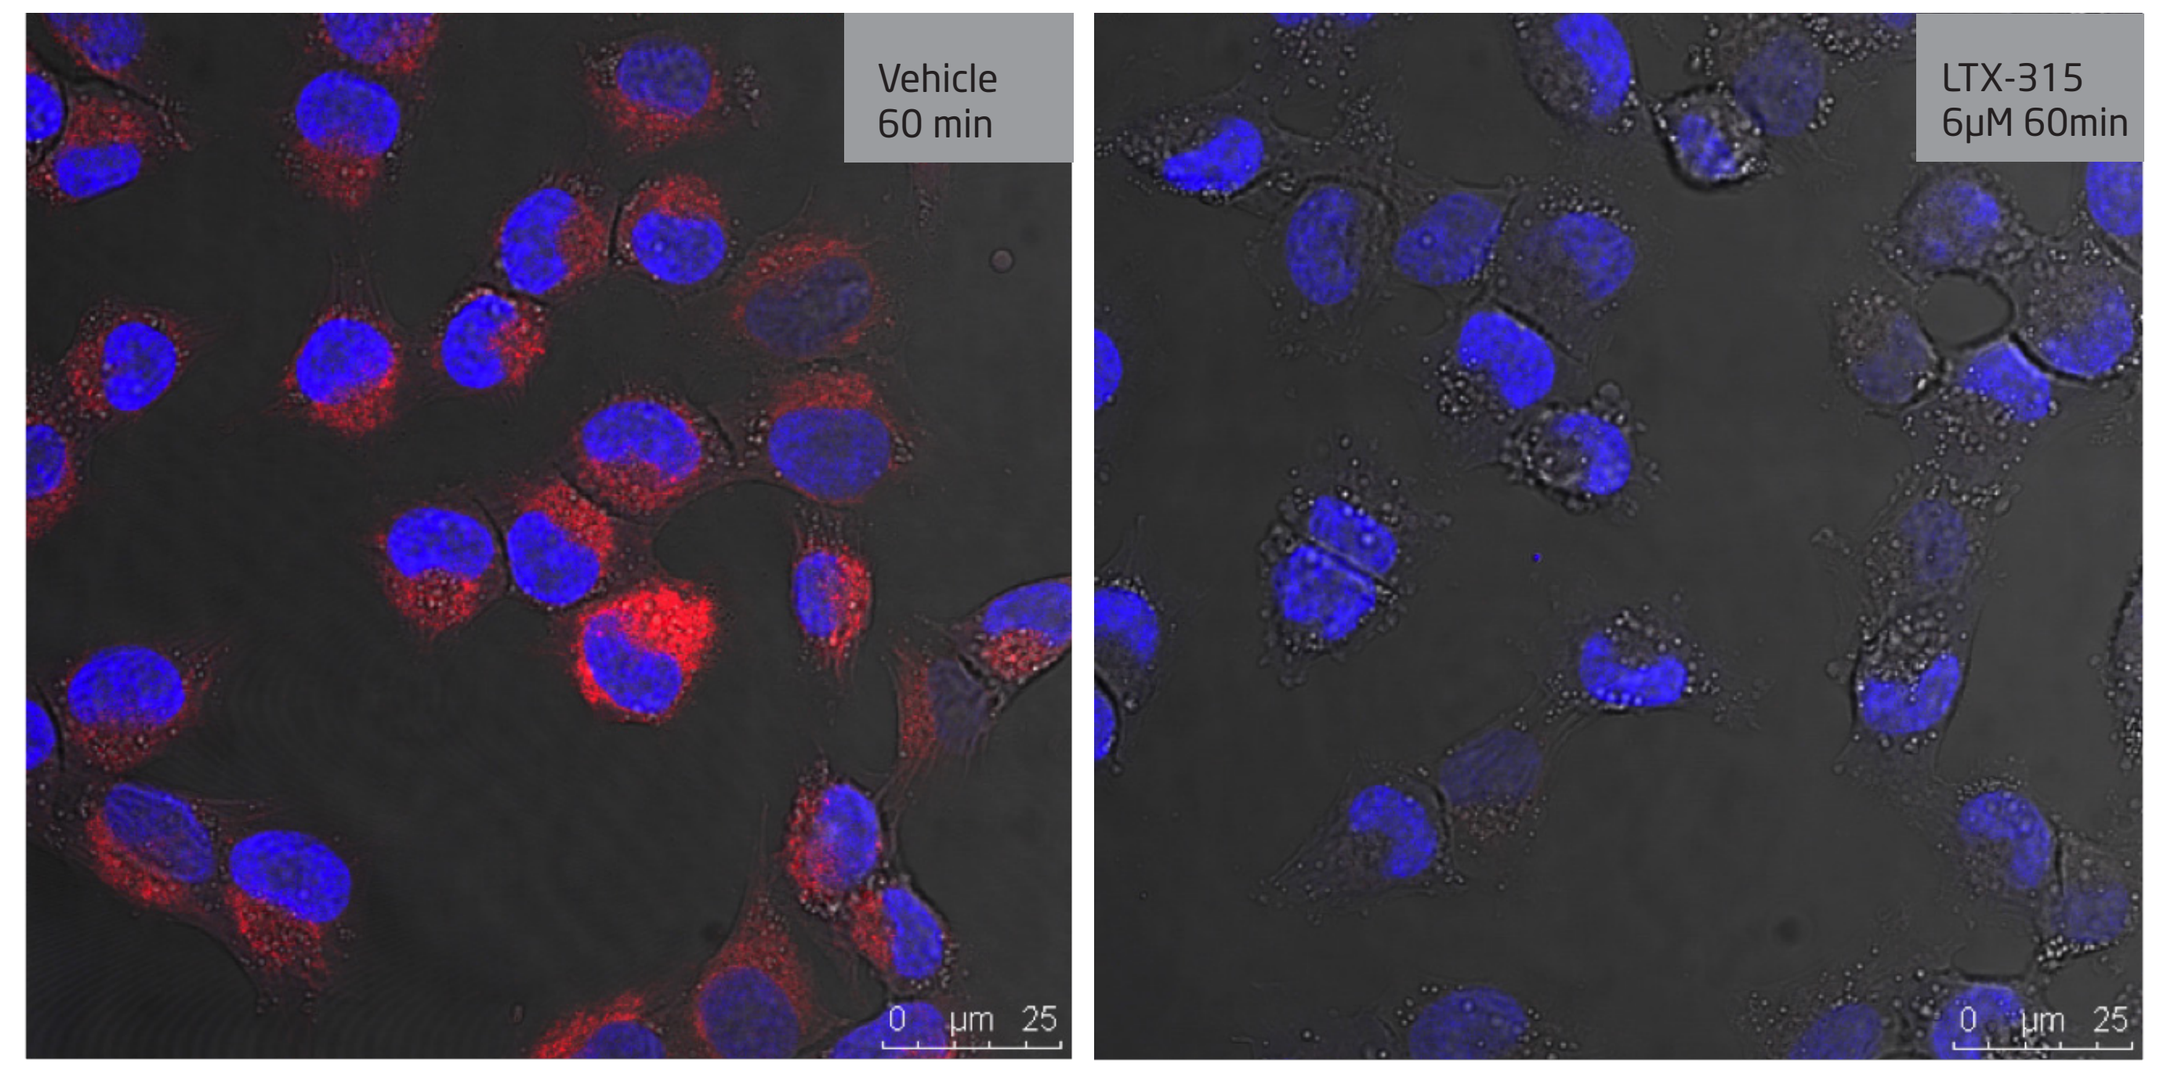
Vehicle
60 min
LTX-315
6µM 60min
0 µm 25
0 µm 25

Untreated A375 cells were labeled with Mitotracker (Red) and nucleus stained with (DAPI) (left). Treatment with LTX-315 caused disintegration of mitochondria (right).

Fig. 6 - LTX-315 induces disintegration of the mitochondria.

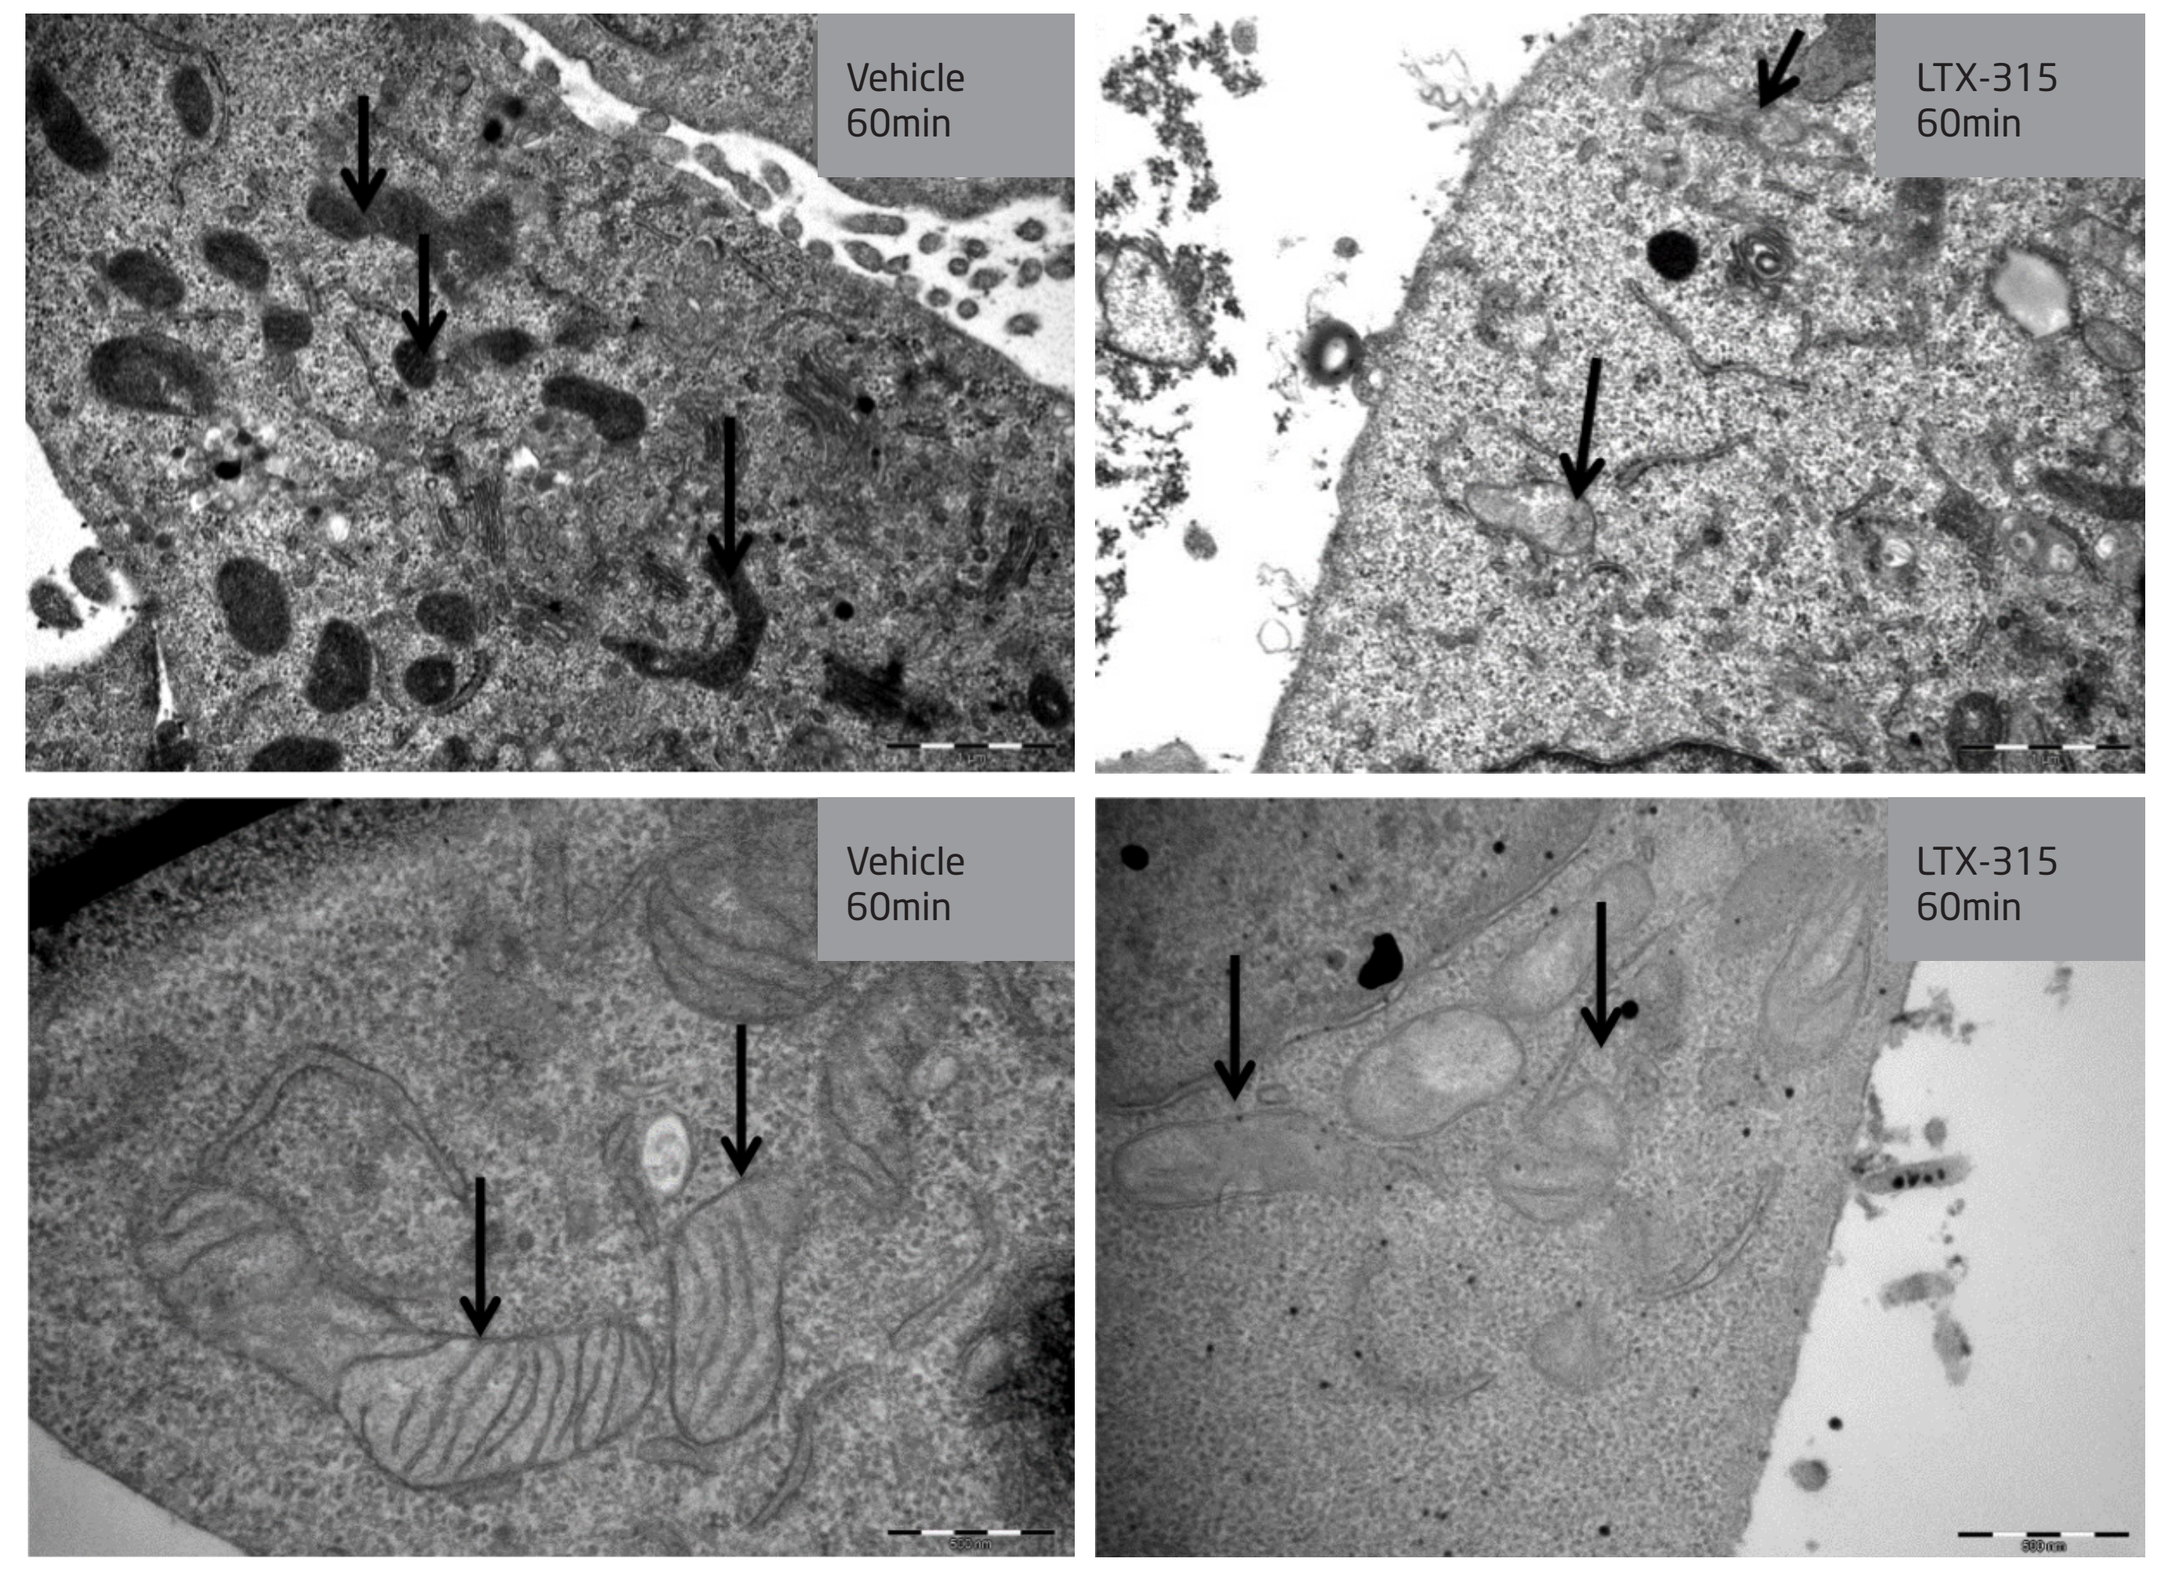
Vehicle
60min
LTX-315
60min
Vehicle
60min
LTX-315
60min

A375 cells were treated with 17µM LTX-315 for 60min and analyzed by transmission electron microscopy (TEM). Arrows: mitochondria.

Fig. 7 - LTX-315 treatment induces cytochrome-C release

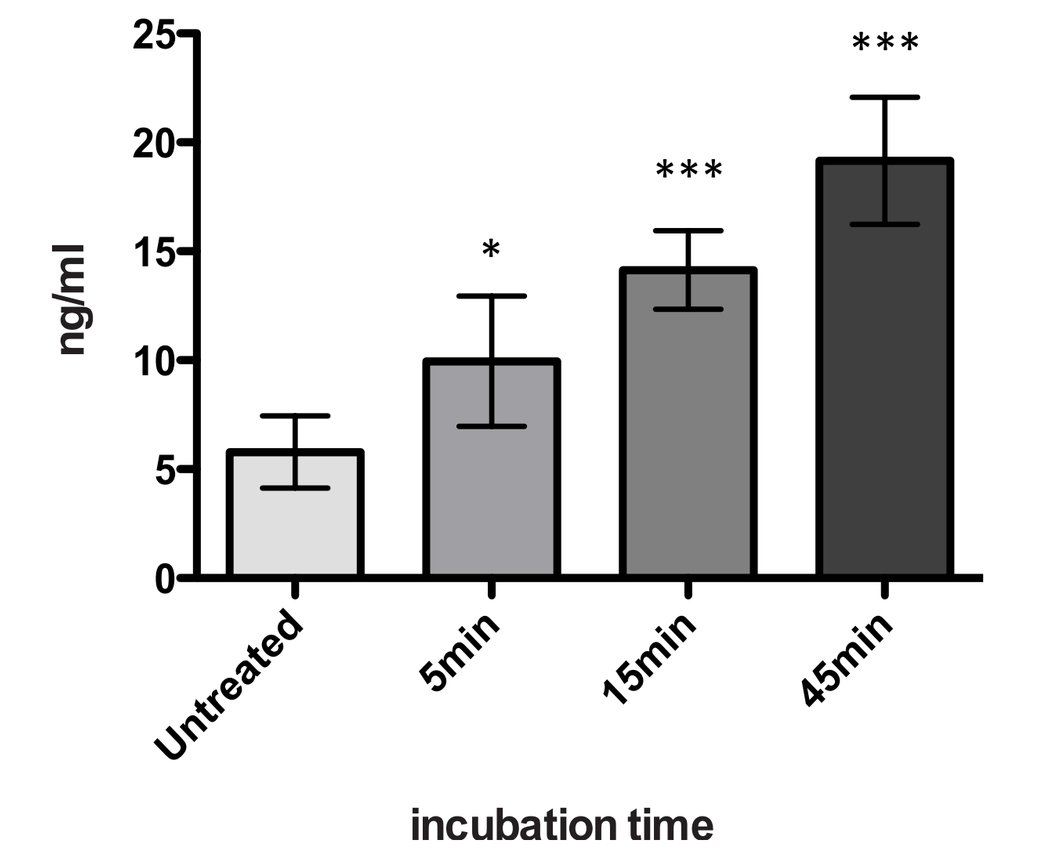
ng/ml
25
20
15
10
5
0
*

Untreated
5min
15min
45min
incubation time

Cytochrome-C release in the supernantant after LTX-315 treatment (35uM) of A375 cell at designated time points (5, 15, 45 min) was determined by ELISA assay.

Fig. 8 - LTX-315 induces Reactive Oxygen Species (ROS)

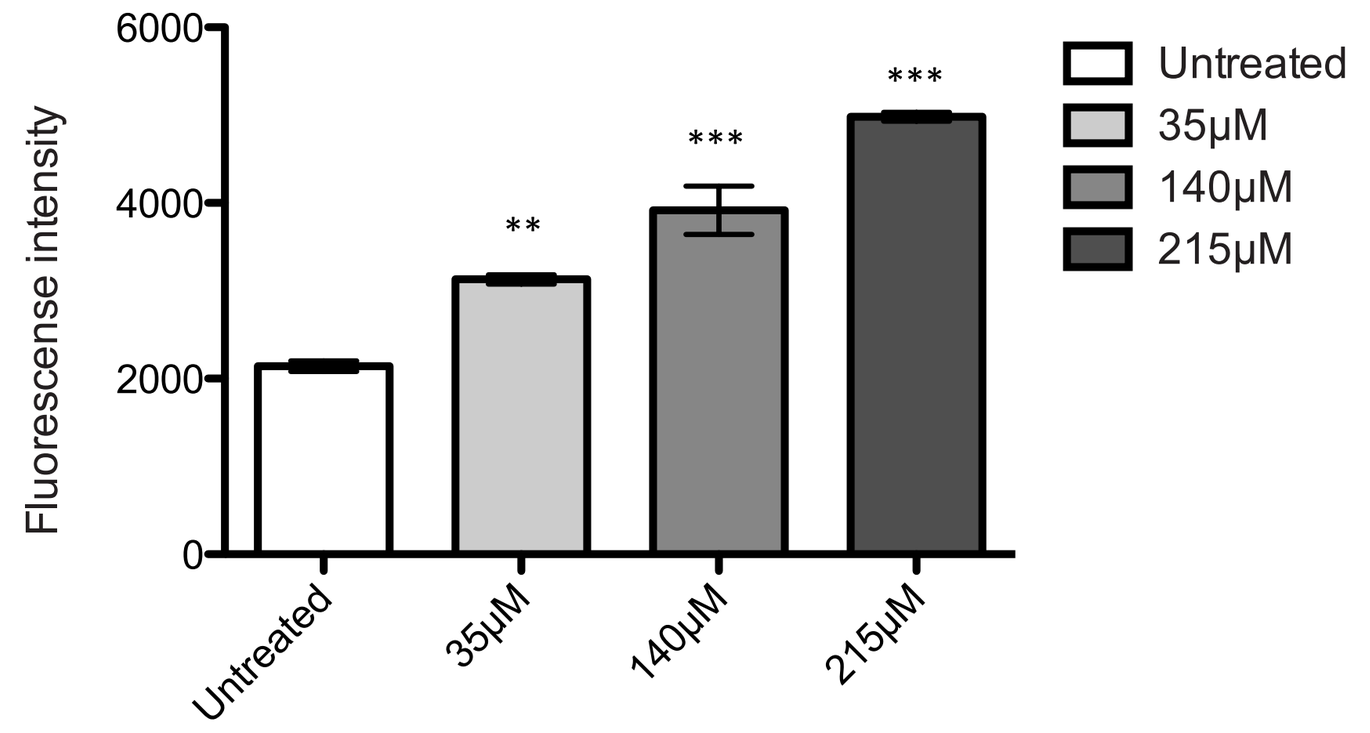
Fluorescense intensity
6000
4000
2000
0
**

Untreated
35µM
140µM
215µM
Untreated
35µM
140µM
215µM

The ROS generation following LTX-315 treatment was measured by fluorometric assay. Significant amounts of ROS were generated after 15 minutes of incubation with LTX-315.

Fig. 9 - Extracellular ATP levels following LTX-315 treatment

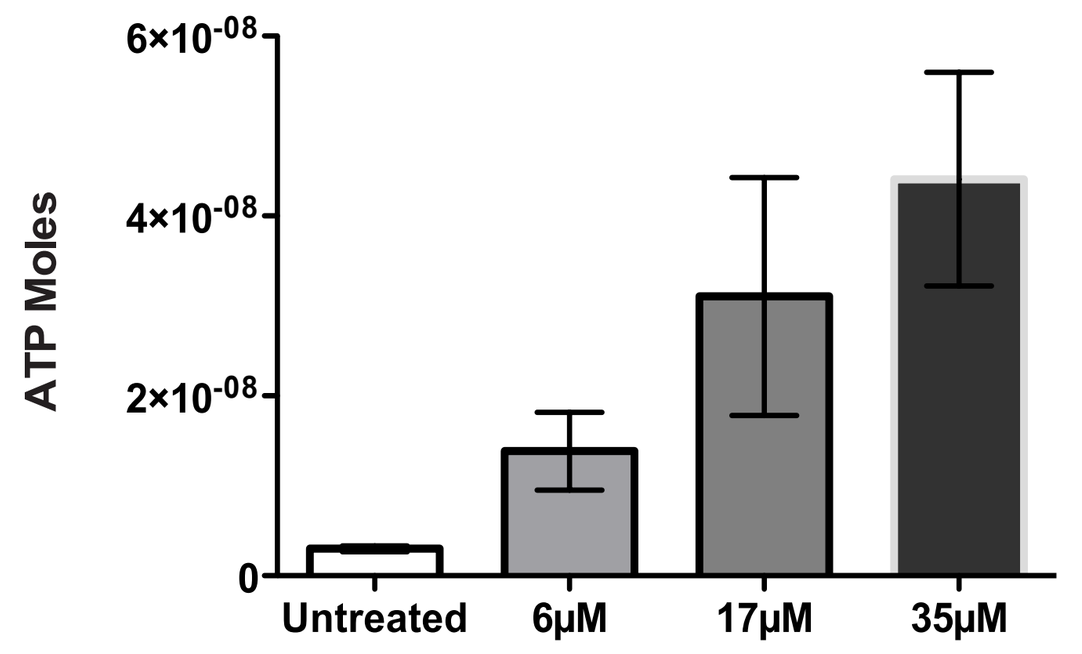
ATP Moles
6×10-08
4×10-08
2×10-08
0
Untreated
6µM
17µM
35µM

A375 cells were treated with LTX-315 for 5 minutes at different concentrations. Quantification of ATP level was performed by luciferase bioluminescence. Results are presented as mean +/- S.D.

Fig. 10 - LTX-315 Induces release of HMGB1

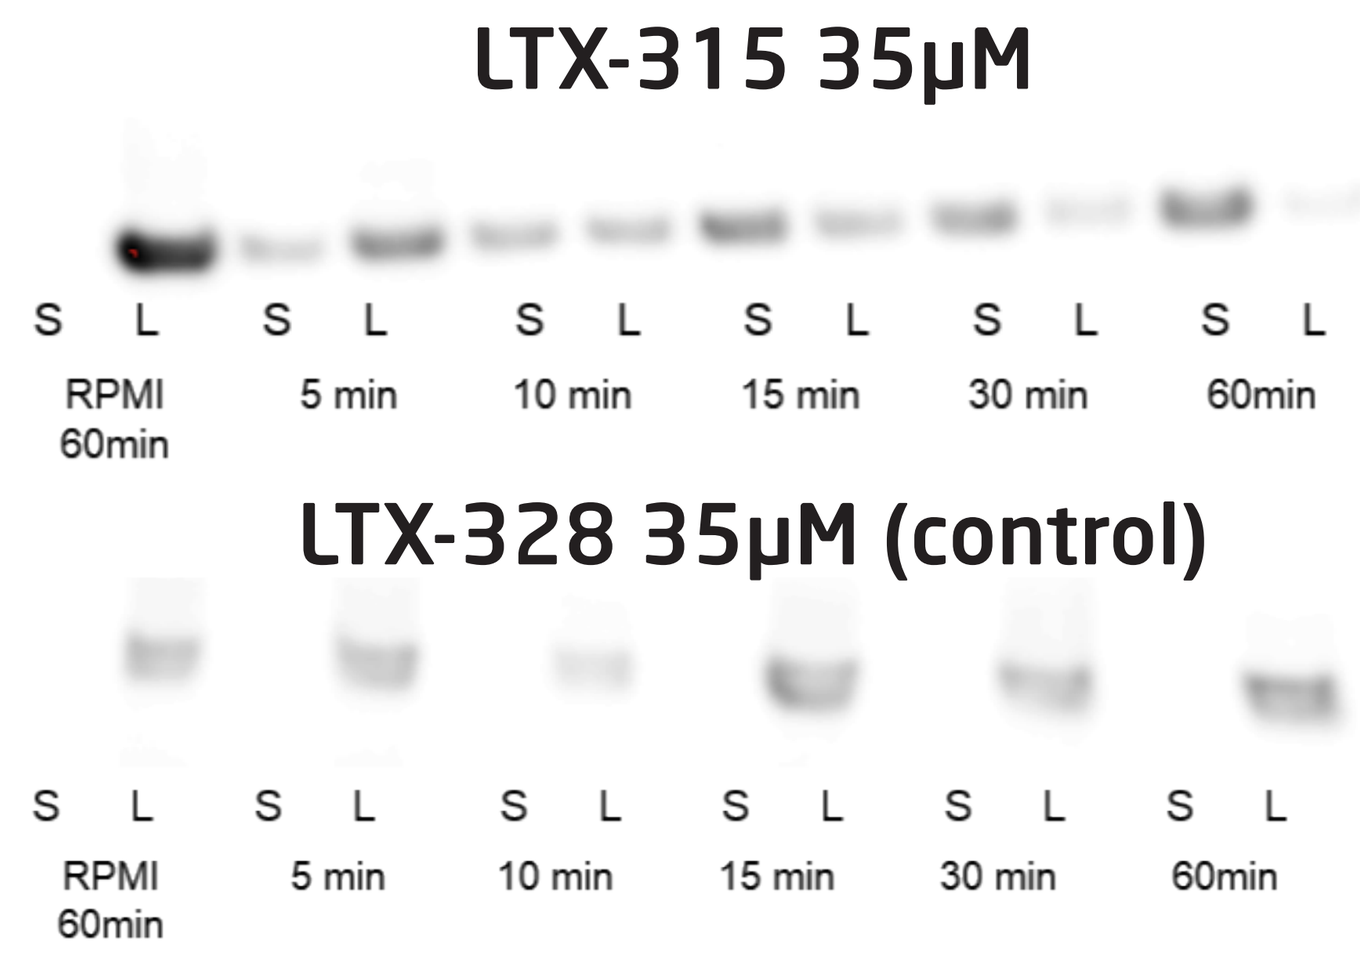
LTX-315 35µM
S L
S L
S L
S L
S L
S L
RPMI 60min
5 min
10 min
15 min
30 min
60min
LTX-328 35µM (control)
S L
S L
S L
S L
S L
S L
RPMI 60min
5 min
10 min
15 min
30 min
60min

LTX-315 Induces release of HMGB1
A375 human melanoma cells were treated with 35 µM LTX-315 (top) or the control peptide LTX-328 (bottom) and cell lysate (L) and supernatant (S) were analyzed with western blot. LTX-315 treated cells showed a gradual translocation from the cell lysate to the cell supernatant. Control cells treated LTX-328 did not showed any translocation of HMGB1 after 60 minutes.

©WWW.CHILIGROUP.NO|2014

Conclusions

These findings demonstrate that LTX-315 has a membrane perturbing effect that results in the release of a number of danger signal molecules (DAMPS). It`s effects against both the cell membrane and the mitochondria membrane may explain LTX-315`s ability to induce complete regression and long term protective immune responses in a number of experimental models (see poster 19 and 20).

Referanser

1. Zasloff, M., Antimicrobial peptides of multicellular organisms. Nature, 2002. 415(6870): p. 389-95.
2. Mader, J.S. and D.W. Hoskin, Cationic antimicrobial peptides as novel cytotoxic agents for cancer treatment. Expert Opin Investig Drugs, 2006. 15(8): p. 933-46.
3. Hoskin, D.W. and A. Ramamoorthy, Studies on anticancer activities of antimicrobial peptides. Biochim Biophys Acta, 2008. 1778(2): p. 357-75.
4. Yang, D., et al., Multiple roles of antimicrobial defensins, cathelicidins, and eosinophil-derived neurotoxin in host defense. Annu Rev Immunol, 2004. 22: p. 181-215.
5. Camilio, K.A., Complete regression and systemic protective immune responses obtained in B16 melanomas after treatmentwith LTX-315. Cancer Immunol. Immunother. 2014 (accepted)

Lytix Biopharma AS | P.O. Box 6447 | NO-9294 Tromsø, Norway | E-mail: post@lytixbiopharma.com | Phone: +47 77 67 55 00 | Fax: +47 77 67 55 01